CHALLENGES WITH MISCONCEPTIONS OF ALOPECIA AREATA AND OPPORTUNITIES TO DRIVE CHANGE: A MULTI-STAKEHOLDER EXPERT PERSPECTIVE

E-Poster number: P664

Pedro Herranz Pinto[1], Jean-Marie Meurant[2], Bianca Maria Piraccini[3], Nisha Pillay[4], Olivia Watkins[4], Ana Maria Sanz[5], Gregor Schaefer[6]

[1]Department of Dermatology, La Paz University Hospital, Madrid, Spain; [2]French Skin Federation, Paris, France; [3]Division of Dermatology, Department of Experimental, Diagnostic and Specialty Medicine, University of Bologna, Bologna, Italy; [4]VML Health, London, UK; [5]Global Public Affairs & Policy, Pfizer; [6]Global Medical Affairs, Pfizer

INTRODUCTION AND OBJECTIVES

- Alopecia areata (AA) is a systemic autoimmune disease that affects up to 2% of the population at some point in their lifetime, with around 60% of patients developing their first patch before the age of 20 years[1]
- AA is characterised by an infiltration of immune cells that disrupt the normal hair growth cycle, resulting in non-scarring hair loss[2]
- Despite its autoimmune nature, AA can be misperceived as merely a cosmetic issue by the public, healthcare professionals (HCPs), payers, and policymakers
- This impedes effective diagnosis and treatment and drives stigma associated with the disease, imposing substantial burden on patients and caregivers and ultimately impacting their quality of life (QoL)[3,4]
- The AA Consensus Task Force, consisting of 45 international multi-disciplinary experts (Table 1), aims to achieve consensus on the challenges associated with AA, clarify the true burden faced by those affected, and identify strategies to improve patient care and outcomes

MATERIALS AND METHODS

Quantitative and qualitative data were collected on the challenges faced by people affected by AA and opportunities to overcome them. Insights were collected from 45 global multi-disciplinary experts from 15 countries (Table 1), forming the Global AA Consensus Task Force, via the following methods:

- Landscape assessment including a targeted literature review (369 abstracts screened, 262 full-texts reviewed, 143 with final extraction), virtual group interviews with five representatives from leading AA and dermatology patient advocacy groups (PAGs), and a review of relevant activity from industry, PAGs and professional associations within AA
- Pre-meeting survey using a mixture of open text, multiple choice, and five-point scale questions shared with Task Force members ahead of the advisory boards
- Virtual advisory boards with Task Force members, consisting of a series of five meetings between April and June 2023 to discuss and align on key challenges and solutions across three cross-cutting areas;
 1. Equity of access to care and treatment in AA
 2. Holistic care and support for patients and caregivers
 3. Multi-stakeholder collaboration

Table 1. Country of residence and area of expertise of AA Task Force Members

	MEDICAL PROFESSIONALS	PSYCHOLOGISTS AND PSYCHIATRISTS	PATIENTS AND PAGS	RESEARCH SOCIETIES
AUSTRALIA	✓	✓	✓	✓
ARGENTINA	✓		✓	
BRAZIL	✓	✓	✓	✓
FRANCE			✓	
GERMANY	✓	✓	✓	✓
HONG KONG	✓			
ITALY	✓		✓	✓
JAPAN	✓		✓	✓
MALAYASIA	✓			
SINGAPORE	✓			
SPAIN	✓	✓	✓	✓
TAIWAN	✓			
TURKEY	✓		✓	
UAE	✓			
USA	✓	✓	✓	✓

UAE – United Arab Emirates; USA – United States of America

RESULTS

The Task Force identified challenges across the AA care pathway, underpinned by a lack of understanding and awareness among various stakeholders. Additionally, they identified opportunities to engage key stakeholder groups in changing the perception of AA and addressing these identified challenges.

CHALLENGE 1
Low public awareness and misconceptions around AA can drive stigma associated with the disease and act as significant barriers to patients presenting to the healthcare system

Many individuals fail to recognise AA as a systemic autoimmune dermatological disease, often misperceiving it as merely a cosmetic condition. Instead of seeking appropriate medical help, many patients turn to non-medical professionals (e.g., beauticians or hair centres), alternative medicine, or other hair loss specialists (e.g., trichologists, hair transplant surgeons), thus delaying diagnosis and adequate treatment. Most [91% (41/45)] experts agreed that a low public awareness of AA may limit understanding and contribute to stigma associated with the disease (Figure 1). Patients often encounter mistaken assumptions from the public regarding the cause of hair loss, such as cancer or stress. Stigma may lead to further delays in seeking help as patients attempt to conceal hair loss or avoid reaching out for support due to fear of judgement, ultimately contributing to a lowered QoL for both patients and caregivers.[3,4]

Figure 1. Expert opinion on public awareness of AA

LOW PUBLIC AWARENESS OF AA CONTRIBUTES TO A POOR UNDERSTANDING OF, AND STIGMA ASSOCIATED WITH THE DISEASE

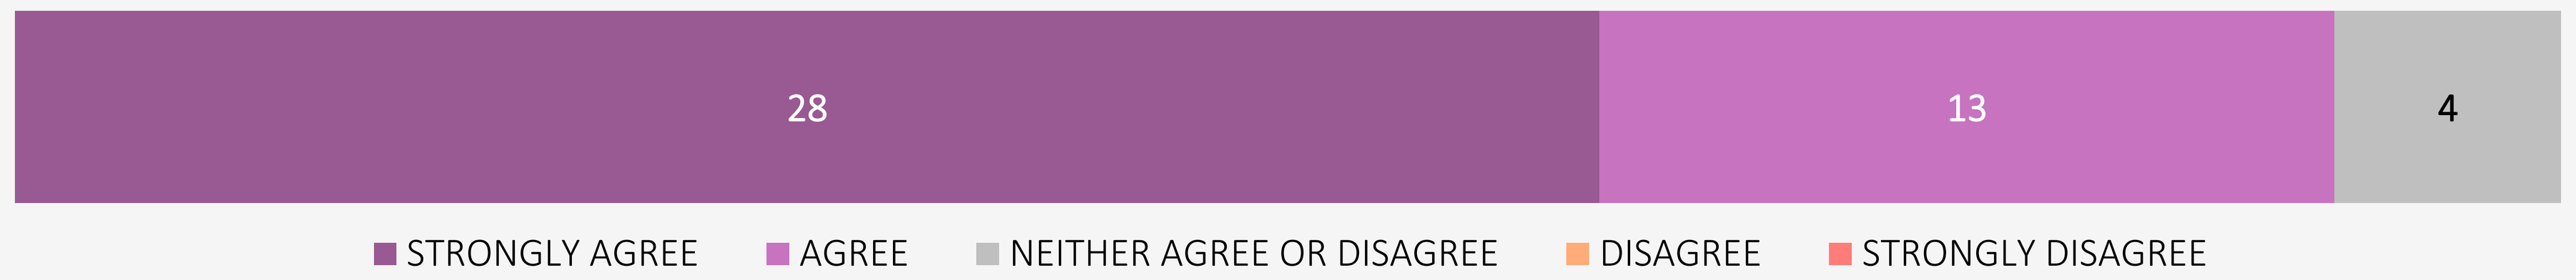
28
13
4
STRONGLY AGREE
AGREE
NEITHER AGREE OR DISAGREE
DISAGREE
STRONGLY DISAGREE

OPPORTUNITY
Improve awareness and understanding of AA among the public to reduce stigma and improve presentation to dermatologists

Create and deliver educational and awareness campaigns to the public:

- Disseminate accurate and accessible information on AA via social media platforms to educate the public on AA and dispel common myths
- Enhance presence at both local and national events (such as sponsored activities, televised events, dedicated awareness months and days) to improve education and awareness among the public
- Leverage key influencers or celebrity ambassadors to increase the visibility of the disease and its burden across wide audiences

CHALLENGE 2
Limited understanding of AA among non-specialist HCPs may act as a barrier to diagnosis and treatment

Professionals lacking the appropriate medical training may struggle to identify hair loss symptoms as indicative of AA, which can result in patients not being directed to the appropriate medical specialists. Even once patients reach a medically trained first-line physician, often primary care physicians (PCPs), these HCPs may lack sufficient awareness and understanding of AA to refer to dermatologists for a formal diagnosis and treatment plan. Most [93% (42/45)] experts noted that many HCPs, especially non-specialists, lack a comprehensive understanding of AA and its holistic impact on patients (Figure 2), which may lead to dismissal of patients.

Figure 2. Expert opinion on HCP understanding of AA

MANY HCPs HAVE A LIMITED UNDERSTANDING OF AA, ITS HOLISTIC IMPACT, AND THE PATIENT EXPERIENCE

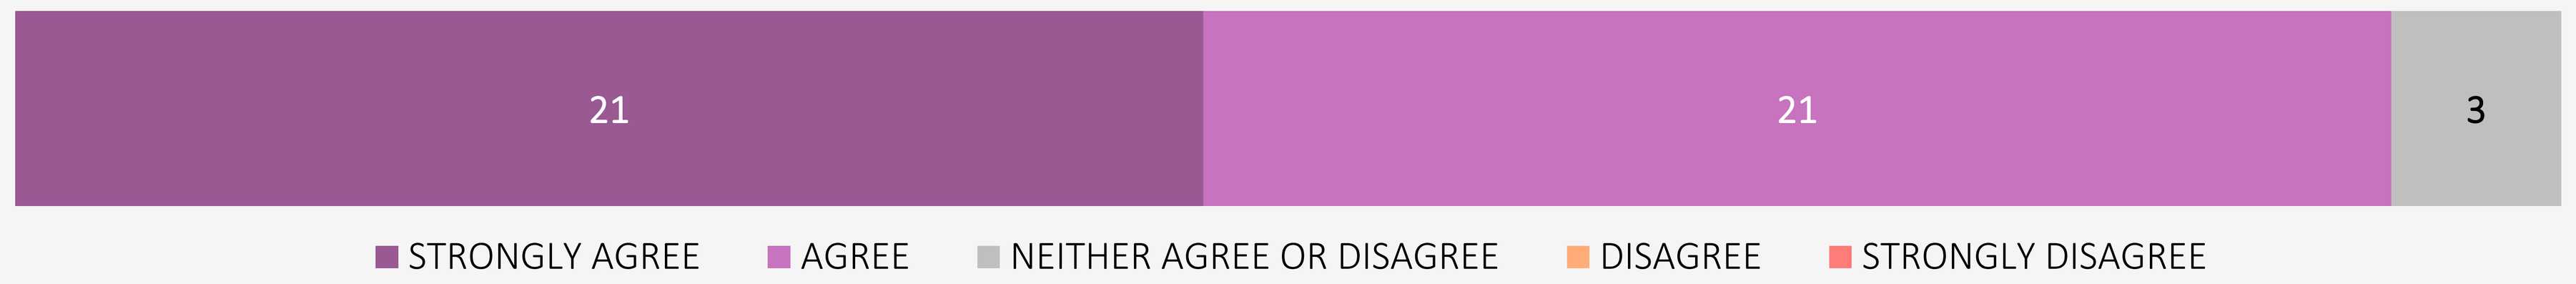
21
21
3
STRONGLY AGREE
AGREE
NEITHER AGREE OR DISAGREE
DISAGREE
STRONGLY DISAGREE

OPPORTUNITY
Educate the non-specialist hair community and PCPs to improve the pathway diagnosis and treatment for patients

Implement initiatives to enhance awareness among professionals and educate PCPs to ensure timely referrals:

- Conduct targeted outreach to increase awareness of AA among hair and skin professionals that may encounter AA patients and encourage them to promptly direct patients to appropriate medical specialists
- Create educational programmes for PCPs to increase understanding of AA and the holistic impact on patients, and improve referral to dermatologists

> “People are ashamed of carrying the disease or explaining what they have, the cancer [misconception] issue is big.”

PATIENT ADVOCATE, ARGENTINA

> “It is important when we talk about stigma to talk about the ignorance about AA, people do not understand the nature and the causes which can contribute to the stigma.”

DERMATOLOGIST, SPAIN

CHALLENGE 3
Lack of recognition of AA as a systemic autoimmune disease by payers and policymakers may prevent reimbursement and limit access to treatment

Due to the non-life-threatening nature of AA, payers and policymakers often overlook the need for treatment as they do not understand the holistic burden experienced by patients, which may lead to limited access to treatment. Most [82% (37/45)] experts agreed that access to AA treatment is low, driven by payer perception, inadequate coverage, and market and regional disparities (Figure 3). This is a particular challenge in certain countries such as Germany, where restrictive policies limit reimbursement for hair loss treatments,[5] and in Latin America, where public insurance coverage for most treatments is limited.[6]

Moreover, access to medical prostheses like wigs or other hair loss cover-up methods is often limited across the countries investigated. Even in cases where partial coverage is available, patients may still face substantial out-of-pocket costs for these methods, exacerbating their financial burden.[7]

Figure 3. Expert opinion on access to treatments

ACCESS TO TREATMENT IS LOW, DRIVEN BY PAYER PERCEPTIONS, INADEQUATE COVERAGE, AND MARKET AND REGIONAL DISPARITIES

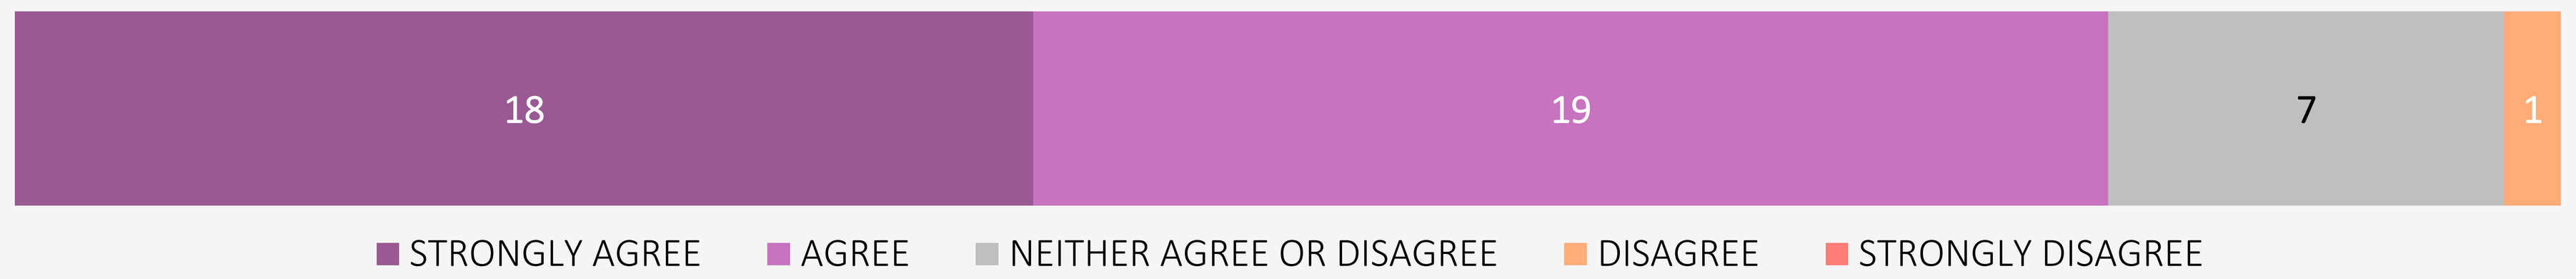
18
19
7
1
STRONGLY AGREE
AGREE
NEITHER AGREE OR DISAGREE
DISAGREE
STRONGLY DISAGREE

OPPORTUNITY
Elevate AA as an autoimmune disease with significant psychosocial burden among all stakeholders, facilitating increased access to comprehensive care

Develop the tools to generate data to illustrate the burden of AA and the potential impact of AA treatments on QoL:

- Conduct patient surveys to assess the true burden of AA and support development of AA-specific psychosocial assessment tools
- Develop new psychosocial assessment tools to be used in clinical practice to accurately measure the holistic burden of AA on patients and caregivers in global populations, expanding on existing assessment tools[8]

Amplify the patient voice:

- Facilitate the presence of patient-centred organisations at large global policy events (e.g., World Health Assembly) to improve the visibility of the disease and its impact
- Encourage the coordination of patient groups to form a stronger patient body which may help elevate the priority of AA on government agendas in regions lacking local PAGs

The three challenges highlighted by the Task Force contribute to the AA care pathway being fragmented. Despite the perception that diagnosing AA is generally straightforward, it is evident that barriers to both diagnosis and treatment represent unmet needs in AA. Most [96% (43/45)] experts agreed that the care pathway is fragmented with multiple barriers to diagnosis and treatment (Figure 4).

Figure 4. Expert opinion on care pathway fragmentation

THE ALOPECIA AREATA CARE PATHWAY IS FRAGMENTED, THERE ARE MULTIPLE BARRIERS TO DIAGNOSIS AND TREATMENT

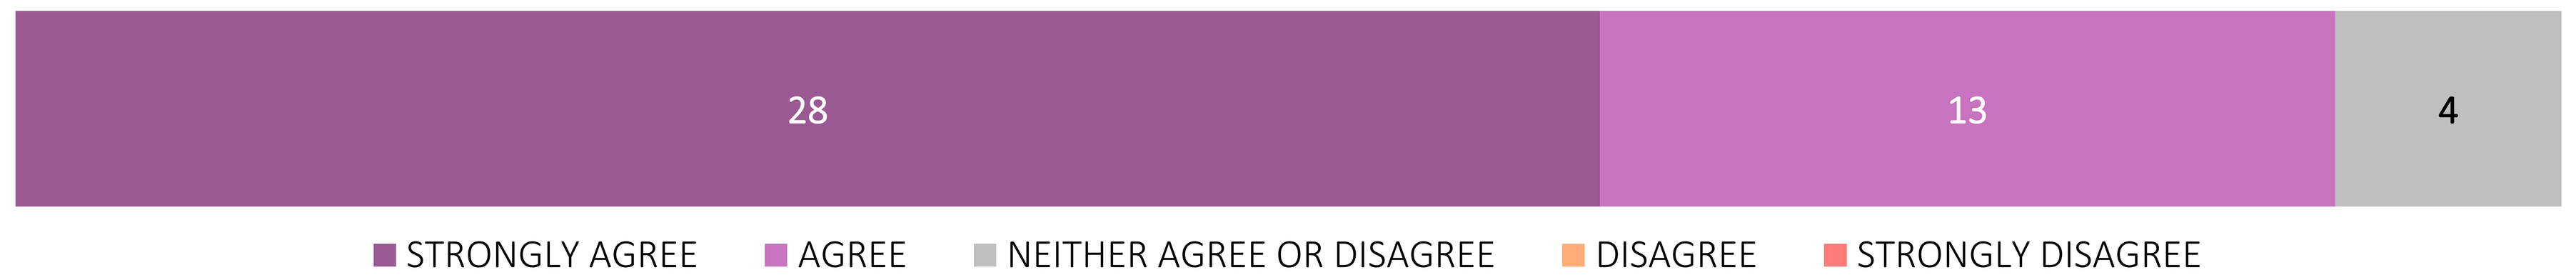
28
13
4
STRONGLY AGREE
AGREE
NEITHER AGREE OR DISAGREE
DISAGREE
STRONGLY DISAGREE

> “Patients who I see have traversed through a tortuous journey to access care.”

DERMATOLOGIST, USA

> “AA is not treated as a disease, for this reason some medical options are not [available] for us.”

PATIENT AND PATIENT ADVOCATE, SPAIN

CONCLUSIONS

Despite AA being an autoimmune disease, persistent misconceptions hinder effective care for those affected. The widespread belief that AA poses only a cosmetic problem to those affected leads to low public awareness, stigma, and limited understanding from HCPs, which in turn impedes diagnosis and treatment. Additionally, payer and policymaker perceptions often result in inadequate coverage and access to treatment for many with AA.

This initiative proposes practical solutions to shift the perception of AA and improve care. Future Task Forces can build on these to enhance outcomes for patients. There is an urgent need for the AA community to work collectively to educate various stakeholders about AA as an autoimmune disease with significant burden on patients and caregivers. Education and increased awareness may help to reduce stigma and initiate progress towards ensuring comprehensive care and support for individuals with AA.

REFERENCES

1. Alkhalifah A, Alsantali A, Wang E, McElwee KJ, Shapiro J. Alopecia areata update: part I. Clinical picture, histopathology, and pathogenesis. J Am Acad Dermatol. 2010;62(2):177–88.
2. Pratt CH, King LE Jr, Messenger AG, Christiano AM, Sundberg JP. Alopecia areata. Nat Rev Dis Primers. 2017;3:17011.
3. Liu LY, King BA, Craiglow BG. Alopecia areata is associated with impaired health-related quality of life: A survey of affected adults and children and their families. J Am Acad Dermatol. 2018;79(3):556-558.e1.
4. Mesinkovska N, King B, Mirmirani P, Ko J, Cassella J. Burden of Illness in Alopecia Areata: A Cross-Sectional Online Survey Study. J Investig Dermatol Symp Proc. 2020;20(1):S62–8.
5. SGB V § 34. Excluded medicines, remedies and aids. Available at: https://www.buzer.de/34_SGB_V.htm. Accessed October 2023..
6. Velásquez-Lopera MM, Hernández N, Jansen AM, García AL, Luna PC, Rico-Restrepo M, et al. Alopecia Areata in Latin America: Where are We and Where are We Going? Dermatol Ther (Heidelb). 2023;13(1):95–114.
7. Ogechi E, Shivali D, Aidan L, J. KK, Lisa A, Nicole F, et al. Patterns of insurance coverage for wigs in patients with alopecia areata: a cross-sectional survey. Int J Womens Dermatol. 2023;9(1):e075.
8. Fabbrocini G, Panariello L, Vita VD, Vincenzi C, Lauro C, Nappo D, et al. Quality of life in alopecia areata: a disease-specific questionnaire. J Eur Acad Dermatol Venereol. 2013;27(3):e276-81.

ACKNOWLEDGEMENTS

The Global AA Consensus Task Force was established and funded by Pfizer and organised by VML Health. We thank all members of the Global AA Consensus Task Force who provided their valuable experience and insights to contribute to this work.

DISCLOSURES

This work was funded by Pfizer and Ana Maria Sanz and Gregor Schaefer are employees. Writing support for this poster was provided by VML Health (London, UK). Other co-authors have no relevant disclosures.